Imputation of missing variance data comparing Bayesian and Classical nonlinear mixed effect modeling to enable a precision weighted meta-analysis

Martin Boucher

Pfizer Research and Development, Sandwich, UK

1. Introduction

When carrying out a meta-analysis of summary level data, the recommended approach is to weight each observation (e.g. arithmetic mean, treatment effect) by its associated precision. Often there are missing standard deviations in the literature and hence an attempt should be made to impute these missing values. The aim was to compare Classical and Bayesian methods of missing data imputation using a non linear mixed effects model.

2. Model and Methods

Internal and external reports were searched to find randomised double blind placebo controlled studies where naproxen had been used to treat subjects with osteoarthritis (OA) pain for knee or hip. The endpoint of interest was mean WOMAC pain score which is typically measured at several time points post dose during a study, the desire being to model the time course for placebo and naproxen. However 30% of the WOMAC pain scores did not have a standard deviation (SD) reported. Figure 1 suggests that an Emax model would provide a good quantitative description of the SD data.

Figure 1 – Standard deviations across time split by treatment and study

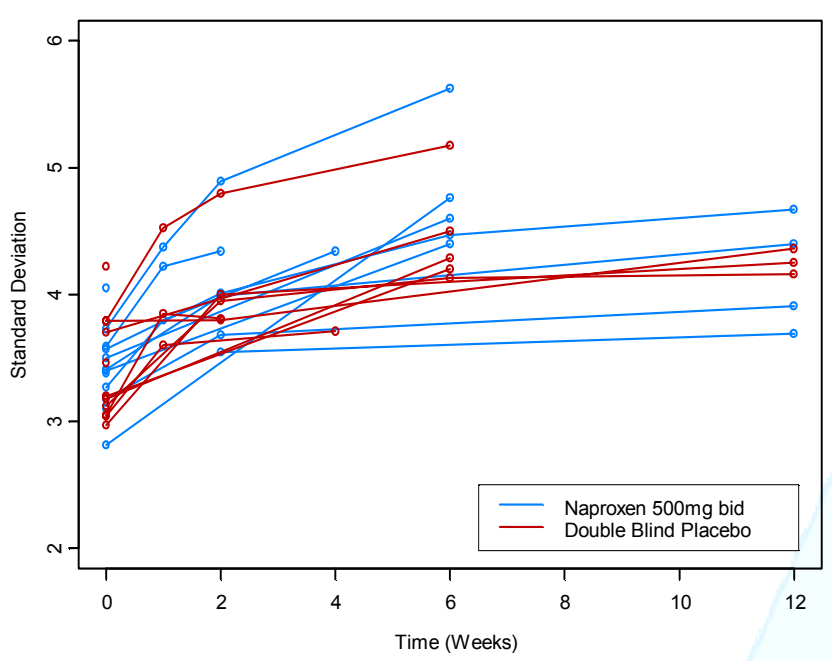
Standard Deviation
2
3
4
5
6
0
2
4
6
8
10
12
Time (Weeks)
Naproxen 500mg bid
Double Blind Placebo

Three approaches were taken to modelling the standard deviations and the WOMAC pain data. These approaches were then compared**.**

The 3 approaches were:

• Using maximum likelihood, fit a longitudinal model to the SDs, predict the missing values and merge these back in to the original dataset. The summary level WOMAC pain data was then modelled separately equating to a 2 stage approach.
• Using Bayesian methodology, fit the same model as the Classical approach and the same 2 stage approach.
• Using Bayesian methods, fit a single model to simultaneously analyse both the standard deviations and the WOMAC pain summary data.

The modelling structure for both the SDs and the summary level WOMAC pain data was as follows (i=study, j=treatment, k=time point)

$$resp_{ijk} = Base + \frac{E_{\max}(p) + I_n * E_{\max}(n)}{te_{50}(p) + I_n * te_{50}(n)} + \eta_i + \varepsilon_{ijk}$$

'Base' is the standard deviation at baseline, Emax(p) is the maximal effect on top of baseline for placebo, I_n is equal to 1 for naproxen data and 0 otherwise and Emax(n) is the additional maximal effect of naproxen over placebo. Emax(n) hence gives an estimate of the treatment effect (naproxen – placebo) at the asymptote.Te_{50} is the time to get to 50% of the maximal effect and is parameterised in the same way as the Emax parameters using the indicator variable I_n. η_i represents the between study variability and is assumed to be distributed $N(0, \sigma^2_b)$. ε_{ijk} is the within study variability and is assumed to be distributed $N(0, \sigma^2_w)$. For the Bayesian modelling, WinBUGS was employed and non informative priors were used for all parameters.

3. Results

Table 1 presents the parameter estimates across the 3 methods. There is some disagreement between the 2 Bayesian approaches with the Emax(n) parameter. There is also a small difference between the Classical and Bayesian methods for the te_{50} parameter.

Table 1 – Parameter estimates for the SD models

	Estimate: Classical (estimate and 95% CI), Bayesian: Median (2.5, 97.5 percentiles of the posterior distribution)		
Parameter	Maximum likelihood	Bayesian model without 2 stage approach	Bayesian model – simultaneous fit
Base	3.4 (3.1, 3.6)	3.4 (3.1, 3.6)	3.4 (3.1, 3.7)
E_{max}(p)	1.4 (1.1, 1.7)	1.4 (1.1, 1.8)	1.4 (1.1, 1.7)
E_{max}(n)	-0.26 (-0.49, -0.01)	-0.26 (-0.51, -0.02)	-0.22 (-0.47, 0.04)
Te_{50} (weeks)	1.3 (0.3, 2.2)	1.5 (0.7, 3.1)	1.5 (0.7, 3.2)
Var(η)	0.12 (0, 0.24)	0.15 (0.06, 0.46)	0.14 (0.03, 0.43)

Figures 2(a) and 2(b) compare the imputed standard deviations from the maximum likelihood approach with the 2 Bayesian approaches. The Bayesian 2 stage approach appears to agree well with the Classical approach (2a) but the agreement is not so strong when comparing to the Bayesian 1 stage approach (2b).

Figures 2 - Predicted SD for missing values comparing the Classical approach with the Bayesian approaches

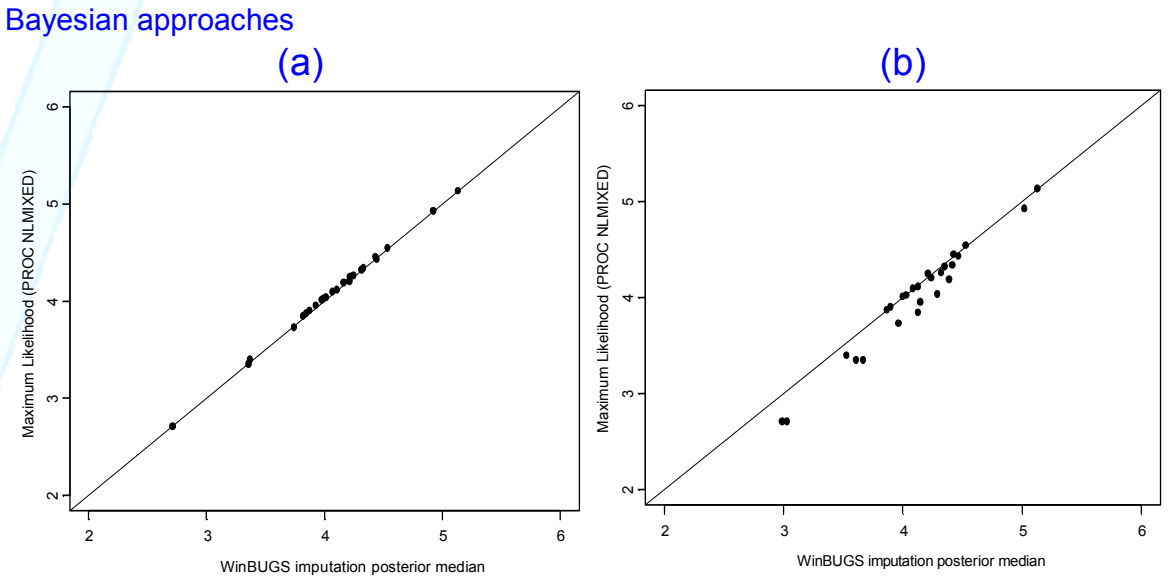
(a)
(b)
Maximum Likelihood (PROC NLMIXED)
WinBUGS imputation posterior median
2
3
4
5
6
Maximum Likelihood (PROC NLMIXED)
WinBUGS imputation posterior median

It was suspected that 'feedback' (the WOMAC pain model influencing the fitting of the SD model) occurring in the joint Bayesian model might be leading to these differences and hence a function in WinBUGS called the 'CUT' function was applied to prevent feedback from the WOMAC pain model to the SD model. This resulted in identical model parameters and predictions between the 2 Bayesian approaches.

4. Discussion/further work

• The reason for this feedback should be investigated further in case it points to some model mis-specification.

• Given that it is sometimes the practice to weight using a function of sample size (hence assuming the SD is constant across time points and studies), it would be interesting to compare such methods with the methods in this paper especially as the assumption of a common SD is not appropriate as demonstrated in Figure 1.

Summary

- A modelling approach was successful in predicting missing standard deviations for a meta-analysis of summary level data.
- The Bayesian 2 stage approach resulted in almost identical imputations as the Classical 2 stage approach.
- The Bayesian 1 stage approach was identical to the 2 stage approach only when feedback was prevented by use of the WinBUGS 'CUT' function.